ASH Annual Meeting
December 9-12, 2017
Abstract 1358

Evaluation of the Correlation between Response and Duration of Treatment in a Phase 2 Study of Pracinostat Plus Azacitidine in Elderly Patients with Acute Myeloid Leukemia (AML)

Ehab Atallah, MD[1], Bruno C Medeiros, MD[2], Samer Khaled, MD[3], Mrinal M. Patnaik, MBBS[4], Riccardo Spezia, MSc[5], Elena Palmesino, PhD[5], and Guillermo Garcia-Manero, MD[6]

[1]Department of Hematology/Oncology, Medical College of Wisconsin, Milwaukee, WI; [2]Division of Hematology-Oncology, Stanford Comprehensive Cancer Center, Stanford University, Palo Alto, CA; [3]Gehr Family Center for Leukemia Research, City of Hope, Duarte, CA; [4]Division of Hematology, Mayo Clinic, Rochester, MN; [5]Helsinn Healthcare, Lugano, Switzerland; [6]Department of Leukemia, The University of Texas MD Anderson Cancer Center, Houston, TX

BACKGROUND

- Inhibition of histone deacetylation and DNA hypermethylation has been shown to induce re-expression of silenced genes in myeloid malignancies in a synergistic fashion[1] and preclinical data suggest that the combination of hypomethylating agents like azacitidine (AZA) with HDAC inhibitors could be synergistic in vivo.[1,2]
- Pracinostat is a potent oral histone deacetylase (HDAC) inhibitor selective for Class I, II, and IV isoforms.
- Pracinostrat in combination with AZA showed promising efficacy in a Phase 2 study in AML patients ≥65 years old not eligible for induction chemotherapy **(Table 1)**.[3]
 - The overall response rate (all complete/partial remissions) was 64%, the complete response (CR) rate was 42%, and the median overall survival was 19.1 months.

Table 1. Previously Reported Efficacy Results (Pracinostat + AZA) in AML

	CR (%)	Median OS (months)
Overall Population (N = 50)	**42.0%**	**19.1**
Age		
66-74 years (N = 24)	**41.7%**	**22.8**
≥75 years (N = 26)	**42.3%**	**15.8**
AML Type		
De Novo (N = 33)	**42.4%**	**13.0**
Secondary (N = 17)	**41.2%**	**29.6**
ECOG Performance Status		
0-1 (N = 43)	**41.9%**	**20.1**
2 (N = 7)	**42.9%**	**13.0**
Cytogenetic Risk Group		
Intermediate (N = 27)	**48.1%**	**24.1**
High (N = 21)	**38.1%**	**13.5**

CR: complete response; **OS:** overall survival

- An evaluation of somatic mutations in this study suggested that continued treatment after achieving a response either lowered the variant allele frequency or cleared residual mutations.[4]
- To further explore the benefit of continued pracinostat treatment we conducted post-hoc analyses to determine the time to first response and assess whether there was a correlation between the duration of treatment and occurrence of a response.

METHODS

Objective

- The objective of this post-hoc analysis was to evaluate whether there was a correlation between the duration of pracinostat treatment and likelihood of a response to treatment.

Study Design and Treatment Groups

- This Phase 2 study evaluated the combination of pracinostat plus AZA in 50 patients diagnosed with AML aged ≥65 years, unfit for induction chemotherapy.
- Pracinostat 60 mg was administered orally 3 days/week (eg, Monday, Wednesday, Friday) for 3 weeks, while AZA 75 mg/m^2 was administered for the first 7 days of each cycle. Treatment cycles were repeated every 28 days.

Key Inclusion Criteria

- Age ≥65 years with newly diagnosed *de novo*, secondary, or treatment-related AML
- Intermediate or unfavorable-risk cytogenetics by US Southwest Oncology Group (SWOG) criteria, ≥20% bone marrow blasts, and Eastern Cooperative Oncology Group (ECOG) performance score (PS) of 0–2

Key Exclusion Criteria

- Acute promyelocytic leukemia, good risk cytogenetics defined as t(15;17), t(8;21), t(16;16), del(16q), or inv(16) karyotype
- Candidate for intensive chemotherapy (induction chemotherapy, bone marrow, or stem cell transplant) within the next 4 months
- Received more than 1 cycle of hypomethylating agent, previous bone marrow transplant, or other intensive chemotherapy for either an antecedent hematologic disorder or AML; received prior HDAC inhibitor or deacetylase inhibitor
- Active central nervous system disease

Efficacy Assessments and Statistical Analysis

- The efficacy endpoint for this analysis was complete response (CR).[5] Responses were assessed at the end of Cycle 2, 4, and 6, and then every 3 cycles until CR was achieved or if there was a suspicion of progressive disease.
- The data cut-off for this study was October 2016, at which point 5 patients continued to receive treatment.
- For this post-hoc analysis, time to CR was calculated and logistic regression modeling was used to assess the probability of a CR utilizing ECOG PS, cytogenetic risk, AML presentation, age class (< /≥75 years), and duration of treatment as covariates. For responders, duration of treatment was censored at the time of occurrence of CR, while duration of treatment for non-responders was the entire time on treatment.
- As it was possible according to the protocol to stop pracinostat or AZA separately, this analysis focused solely on the duration of pracinostat treatment. All 50 patients were included in the analysis.

RESULTS

Baseline Characteristics

- The median age of patients was 75 years. Most patients were white males with *de novo* AML, ECOG PS 0-1, and with intermediate risk cytogenetics. Median bone marrow (BM) blasts at entry were 40% (range 20%-89%).

Table 2. Baseline Demographics and Disease Characteristics

Characteristic	Pracinostat + AZA N = 50
Sex	
Male	**29** (58%)
Female	**21** (42%)
Age (years)	
Median (range)	**75** (66 – 84)
No. (%) Age ≥ 75 years	**26** (52%)
Race	
White	**42** (84%)
Black	**3** (6%)
Asian	**3** (6%)
Unknown	**2** (4%)
ECOG Performance Status	
0-1	**43** (86%)
2	**7** (14%)
AML Presentation	
De novo	**33** (66%)
Secondary to MDS, MPN, or prior chemotherapy/radiotherapy	**17** (34%)
Bone Marrow Blasts	
Median (range)	**40%** (20% - 89%)
Cytogenetic Risk Group	
Intermediate	**27** (57%)
Cytogenetically normal	**21** (42%)
Cytogenetically abnormal	**6** (12%)
Poor*	**21** (42%)
Not classified	**2** (4%)

*Poor risk defined using SWOG definition, and included: Del(5q)/-5, -7/del(7q), abnormal 3q, 9q, 20q, 17p, t(6;9), t(9;22) and complex karyotypes (≥ 3 unrelated abnormalities)

Treatment Cycles

- The median number of treatment cycles was 6.5 (range 1-27+) **(Table 3)**. While 27 patients (54%) received ≥5 cycles, 13 (26%) received only 1-2 cycles.

Table 3. Treatment Cycles/Pracinostat Exposure

Characteristic	Pracinostat + AZA N = 50
Number of Cycles Initiated	
Mean (SD)	**8.5** (7.25)
Median	**6.5**
Minimum, Maximum	**1, 27**
1-2	**13** (26%)
3-4	**10** (20%)
≥5	**27** (54%)
Number of Days on Pracinostat	
Mean (SD)	**259.3** (236.85)
Median	**196.5**
Minimum, Maximum	**5, 904**

Correlation between Time on Treatment and Response to Treatment

- Median time to bone marrow blasts <5% was 2 cycles (57 days; range 25-243); however, 3 patients (12%) required >6 cycles to respond.
- Of the 21 patients achieving a CR, 15 (71%) achieved this after at least 100 days of therapy. **Figure 1** depicts the cumulative incidence of CR by time on treatment.

Figure 1. Cumulative Incidence of CR

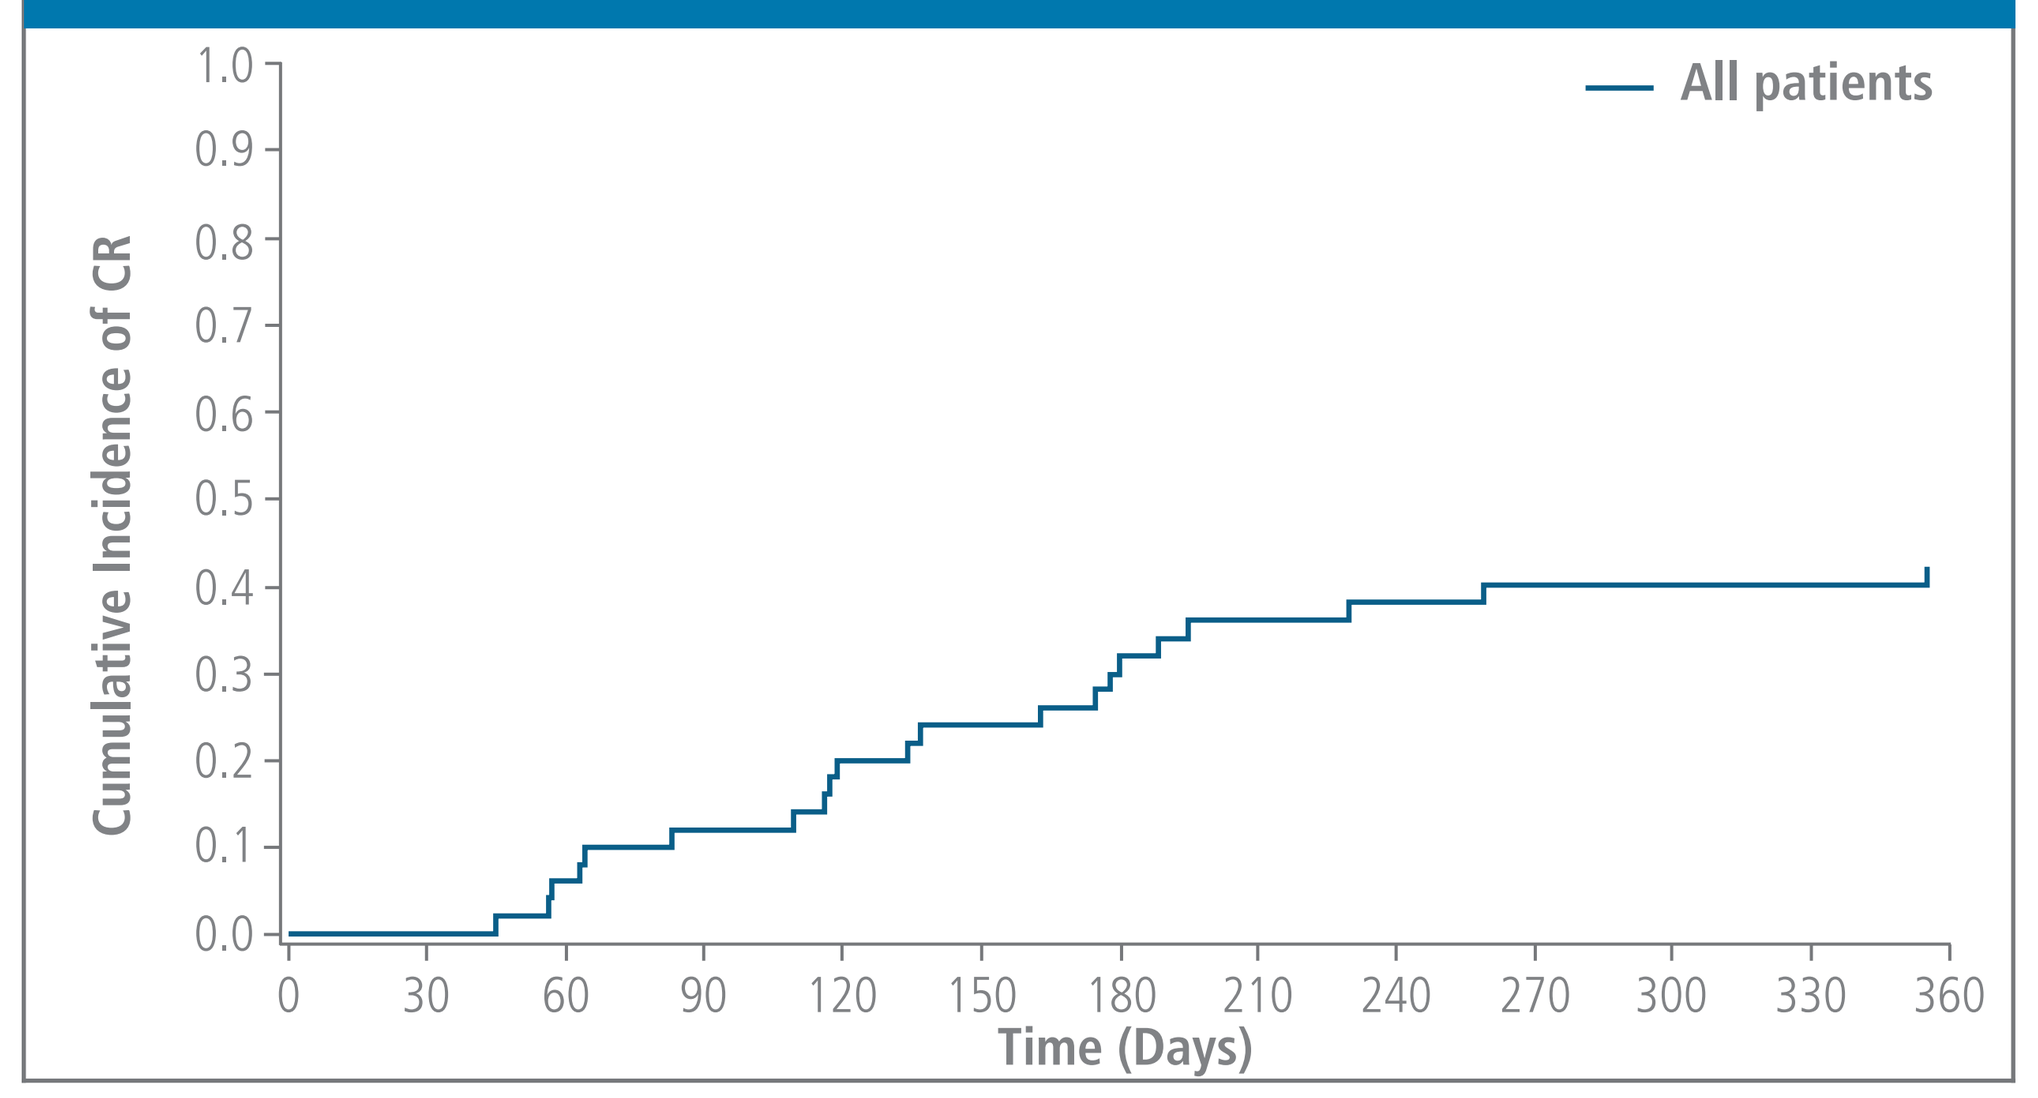
All patients
Cumulative Incidence of CR
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
30
60
90
120
150
180
210
240
270
300
330
360
Time (Days)

- In the subset of patients who responded to pracinostat treatment, the mean time to a CR (by IWG criteria) was approximately 5 cycles, while the mean total time on treatment for those who did not respond was just 3 cycles.
- The mean duration of treatment in the responder subset was 16 months.
- No patient had a CR after having stopped treatment.
- **Figure 2** depicts the median time to and duration of treatment in responders and non-responders.

Figure 2. Median Time to and Median Duration of Pracinostat Treatment in Patients who Responded to Treatment vs Those Who Did Not

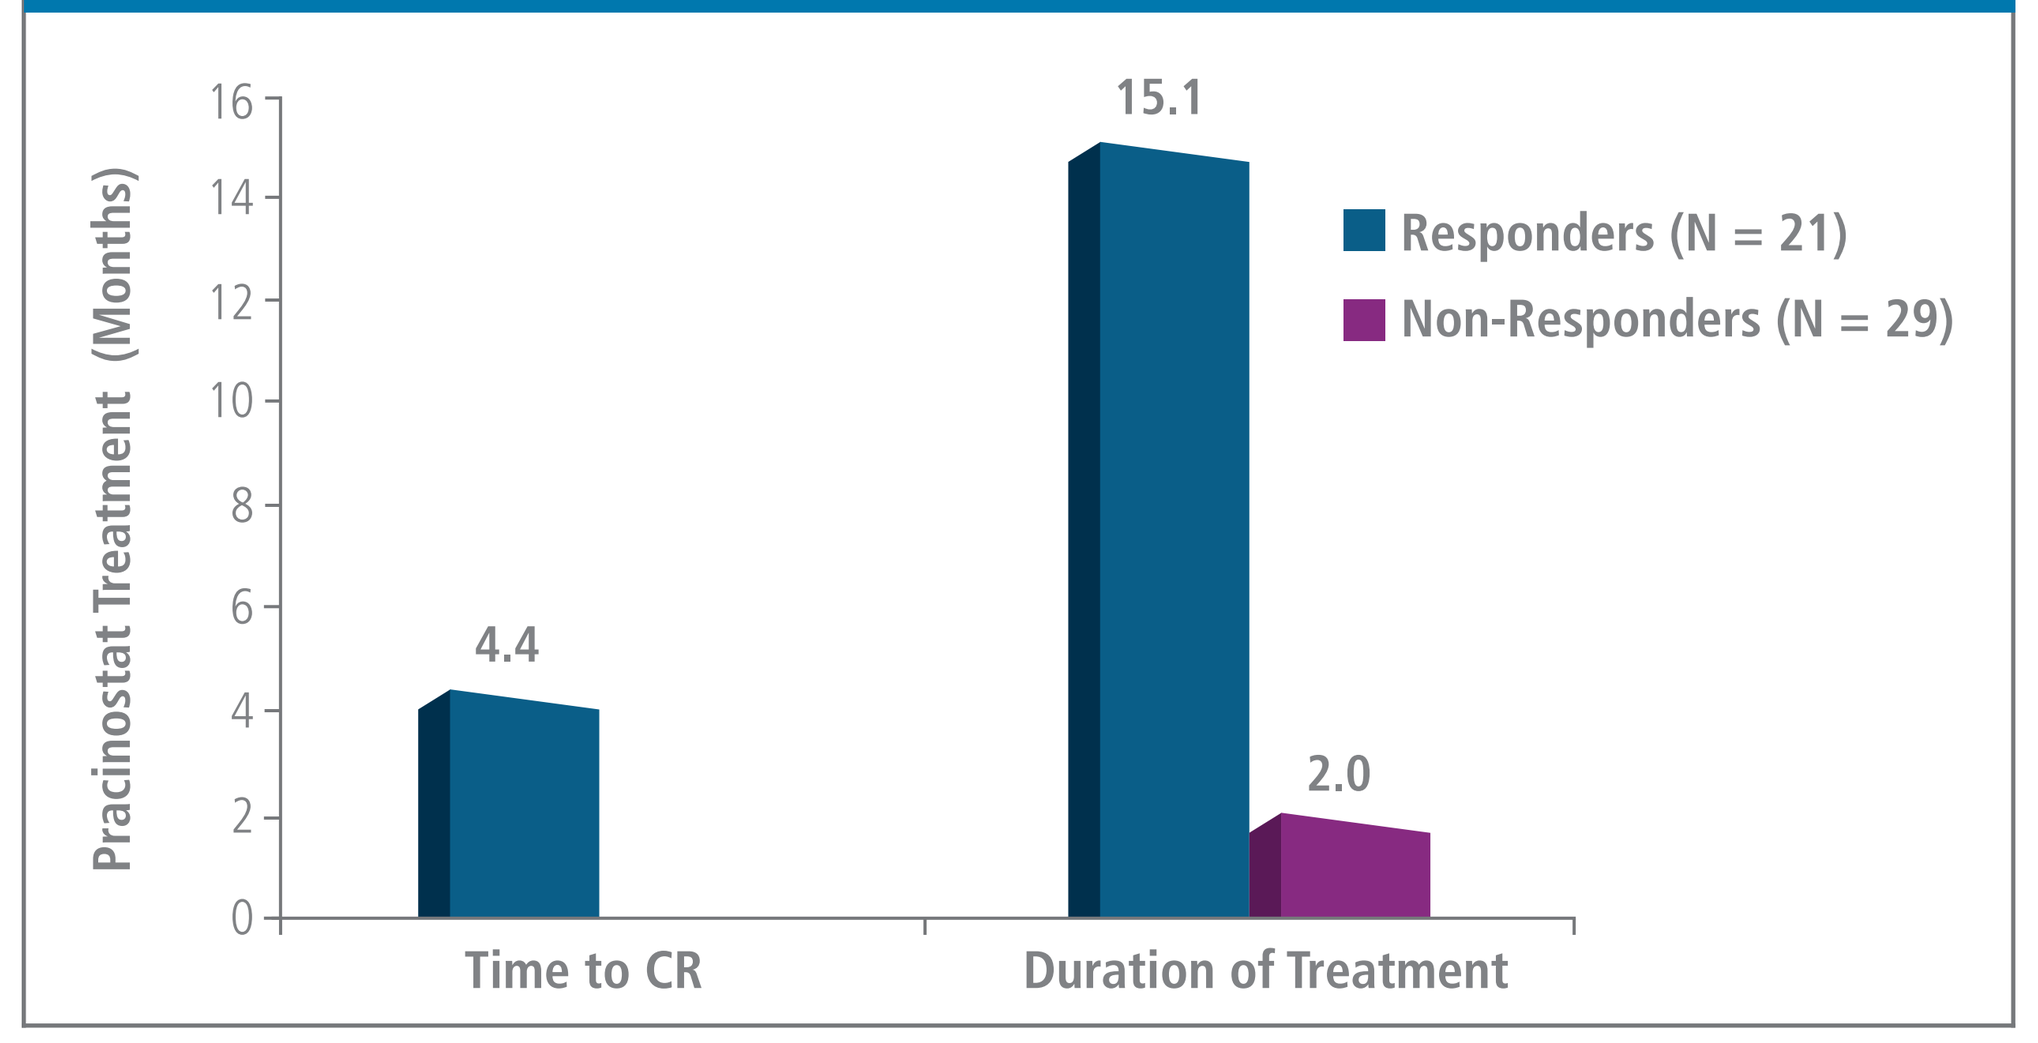
Pracinostat Treatment (Months)
16
14
12
10
8
6
4
2
0
4.4
15.1
2.0
Responders (N = 21)
Non-Responders (N = 29)
Time to CR
Duration of Treatment

- The logistic regression analysis indicated a statistically significant correlation between treatment duration and occurrence of CR **(Figure 3)**.

Figure 3. Regression Line (and 95% CI) between Duration of Treatment (Months) and Probability of Response

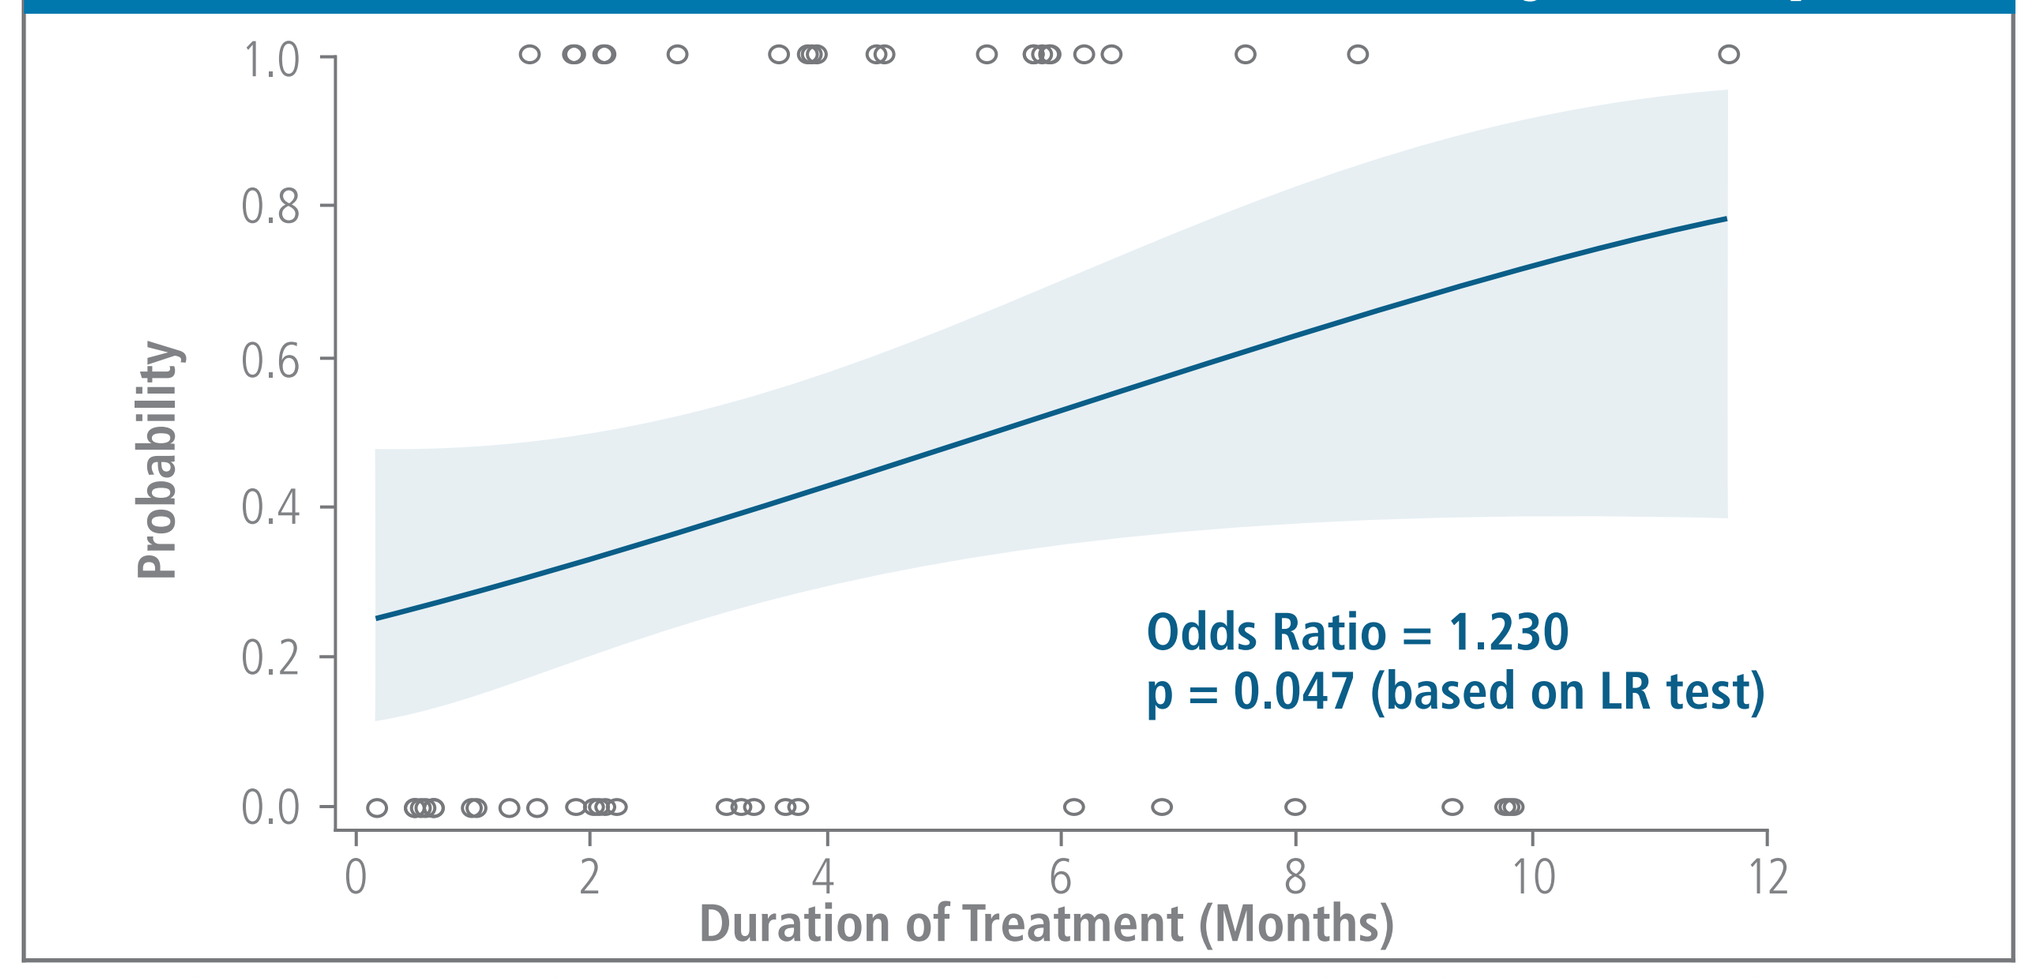
Probability
1.0
0.8
0.6
0.4
0.2
0.0
Odds Ratio = 1.230
p = 0.047 (based on LR test)
0
2
4
6
8
10
12
Duration of Treatment (Months)

Shaded area=95% CI; data points on top of graph=responders; data points on bottom of graph=non-responders

- All patients achieving CR were treated at least up to clinical response and generally low variability in terms of treatment duration was present in the data; some weakness in the inferential results was expected.
- Similar results (P=0.060, Odds Ratio=1.246) were obtained when prognostic factors (ie, ECOG PS, cytogenetic risk, AML presentation, age class (</≥75 years) and duration of treatment) were included in the model.
- It is noteworthy that 9 patients survived less than 3 months and never responded to treatment; this early mortality may potentially introduce bias in this evaluation.
- When the analysis was performed excluding these 9 patients who died within the first 3 months, although a trend was visible, the correlation was not statistically significant (P=0.4681, Odds Ratio=1.087).

Safety

- With a median follow-up of 22.8 months (range 1-31.0), 19 patients (38%) discontinued due to progressive disease.
- Eleven (22%) patients discontinued due to patient or investigator decision and 14 (28%) due to adverse events. The majority of these adverse events were fatigue, failure to thrive, neutropenia, or thrombocytopenia.
- Of the patients discontinuing due to adverse events, 6 discontinued during the first 3 cycles, 2 between cycles 4-6, and the remaining 6 after cycle 6.
- No unexpected toxicities were observed with the combination of pracinostat and AZA.

CONCLUSIONS

- A high response rate with pracinostat + AZA has been previously reported in this study of elderly patients with AML.[3]
- This post-hoc analysis suggests that while marrow remission may be achieved relatively early, prolonged exposure to pracinostat treatment is likely to be beneficial in maximizing the likelihood of a complete response with hematologic recovery.
 - This is consistent with the prior evaluation of somatic mutations that suggested that continued treatment after achieving a response either lowered the variant allele frequency or cleared residual mutations.[4]
- A phase 3 study of pracinostat in combination with AZA in untreated patients with AML unfit for standard induction chemotherapy is ongoing.
 - In line with these findings, it is recommended that patients in that study who have no evidence of disease progression should receive a minimum of 6 cycles of study treatment, if and when possible, to maximize the likelihood of a response to treatment.

REFERENCES: 1. Cameron EE, et al. Nat Genet 1999;21:103-107. **2.** Schneider-Stock R, Ocker M. IDrugs 2007;10:557-561. **3.** Garcia-Manero G, et al. Blood 2016;128:100. **4.** Takahashi K, et al. J Clin Oncol 2017;35:(suppl; abstr 7034). **5.** Cheson BD, Bennett JM, Kopecky KJ, et al. J Clin Oncol 2003;21:4642-4649.

Author Conflicts of Interest: Ehab Atallah: research funding (ADC Therapeutics); Samer Khaled: travel support (Daiichi Sankyo, Inc); research funding (City of Hope); Riccardo Spezia: employee, patents and royalties (Helsinn Healthcare); Elena Palmesino: employee (Helsinn Healthcare); Mrinal Patnaik, Bruno Medeiros, and Guillermo Garcia-Manero: nothing to disclose

ACKNOWLEDGEMENTS: The authors would like to thank the clinical investigators, patients and site personnel at each of the participating centers. Editorial and medical writing assistance was provided by Jennifer Vanden Burgt, Minneapolis, MN and funded by Helsinn Healthcare SA, Lugano, Switzerland. The trial was sponsored by Helsinn Healthcare SA.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

For questions, contact Dr. Atallah at eatallah@mcw.edu