Treatment of Pracinostat and Azacitidine in Elderly Patients With Acute Myeloid Leukemia (AML): Correlation Between Mutation Clearance and Clinical Response

22nd EHA Congress
June 22–25, 2017
Madrid

Koichi Takahashi,[1] Yasmin Abaza,[1] Feng Wang,[1] Curtis Gumbs,[1] Song Xingzhi,[1] Andrew Futreal,[1] Ehab Atallah,[2] Bruno C. Medeiros,[3] Samer K. Khaled,[4] Martha Arellano,[5] Mrinal M. Patnaik,[6] Elena Palmesino,[7] Guillermo Garcia-Manero[1]

[1]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Department of Hematology/Oncology, Medical College of Wisconsin, Milwaukee, WI, USA; [3]Department of Medicine, Division of Hematology, Stanford University, Stanford, CA, USA; [4]Department of Hematology and Hematopoietic Cell Transplantation, and Gehr Family Center for Leukemia Research, City of Hope, Duarte, CA, USA; [5]Emory University School of Medicine, Division of Hematology, Atlanta, GA, USA; [6]Mayo Clinic, Division of Hematology, Rochester, MN, USA; [7]Helsinn Healthcare, Lugano, Switzerland

BACKGROUND

- Older patients with acute myeloid leukemia (AML) have poor prognosis, with a median overall survival (OS) of 5–10 months[1]
 - A greater understanding of the genetic abnormalities and their association with patient outcomes, as well as more effective and better-tolerated treatments, are needed for AML
- In a phase 3 study in AML patients aged ≥65 years who were not candidates for intensive therapy and stem cell transplant, the hypomethylating agent azacitidine resulted in a complete-response (CR) rate comparable to that achieved with conventional care regimens (19.5% vs 21.9%) and a trend for better median OS (10.4 months vs 6.5 months) and 1-year OS (46.5% vs 32.4%)[2]
- Pracinostat is an investigational, oral hydroxamic acid-based histone deacetylase (HDAC) inhibitor selective for classes I, II, and IV isoforms
 - HDAC inhibitors and azacitidine have demonstrated synergistic activity in vitro[3]
- In a single-arm phase 2 study in 50 patients aged ≥65 years with previously untreated AML who were not eligible for intensive induction chemotherapy, pracinostat and azacitidine led to 42% CR rate and a median OS of 19.1 months[4]
 - Responses were durable (median CR+CR with incomplete blood recovery [CRi] 17.2 months)
 - Blast clearance was rapid (median 8 weeks)
 - Maximum clinical benefit required prolonged therapy (>6 months) in some patients

OBJECTIVE

- This post-hoc analysis of the phase 2 study was conducted to investigate the impact of somatic mutations and their clearance on survival outcomes in patients treated with pracinostat and azacitidine

METHODS

Study Design

- The phase 2 study evaluated the combination of pracinostat and azacitidine in 50 AML patients aged ≥65 years
 - Pracinostat 60 mg administered orally 3 days/week (eg, Monday, Wednesday, Friday) for 3 weeks, and azacitidine 75 mg/m^2 administered daily intravenously/subcutaneously for 7 days. Treatment cycles were repeated every 28 days
- Primary endpoint: CR+CRi+morphologic leukemia-free state (ie, marrow CR)
 - Responses were assessed at the end of Cycle 2, 4, and 6, and then every 3 cycles until CR achieved or if there was a suspicion of progressive disease
- Secondary endpoints: Overall response rate (ORR); cytogenetic CR; duration of response; progression-free survival; OS; safety and tolerability profile

Patient Population

- Key inclusion criteria: Age ≥65 years; newly diagnosed *de novo*, secondary, or treatment-related AML; intermediate or unfavorable-risk cytogenetics by US Southwest Oncology Group criteria; ≥20% bone marrow blasts; Eastern Cooperative Oncology Group (ECOG) performance score 0–2; informed consent
- Key exclusion criteria: Acute promyelocytic leukemia (FAB M3); good risk cytogenetics defined as t(15;17), t(8;21), t(16;16), del(16q), or inv(16) karyotype; candidate for intensive chemotherapy (induction chemotherapy, bone marrow, or stem cell transplant) within the next 4 months; received more than 1 cycle of hypomethylating agent, previous bone marrow transplant, or other intensive chemotherapy for either an antecedent hematologic disorder or AML; received prior HDAC inhibitor or deacetylase inhibitor; active central nervous system disease

Molecular Mutation Analyses

- 41 of 50 patients had a pre-treatment bone marrow sample available for analyses and 19 of these 41 patients had serial bone marrow samples (median=3) obtained while on therapy
- A total of 88 samples were sequenced
- Leukemia mutations were detected by SureSelect targeted capture exon sequencing (Agilent) of 295 genes that are recurrently mutated in hematologic malignancies (median coverage 507× [range: 111–777×])
- High-confidence somatic mutations were called using modified version of Mutect and Pindel algorithm
- Longitudinal mutation clearance was analyzed by plotting variant allele frequency (VAF) over time
- Association with mutations and treatment response was analyzed by Fisher exact test or Chi-square test, whichever is appropriate
- Survival was analyzed by Kaplan-Meier method, and difference in survival based on mutations was tested by log-rank test

RESULTS

- Demographics and disease characteristics were comparable in the total study population and the gene sequencing population as shown in **Table 1**

Table 1. Baseline Demographics and Disease Characteristics

	Total Study Population (N=50)	Gene Sequencing Population (n=41)
Age		
Median (range), years	**75** (66–84)	**74** (66–84)
No. (%) age ≥ 75 years	**26** (52%)	**20** (49%)
Bone Marrow Blasts Median (range)	**40%** (20–89%)	**40%** (20–84%)
Gender, Male	**29** (58%)	**24** (59%)
ECOG Performance Status 0–1	**42** (84%)	**37** (90%)
AML Presentation		
De novo	**33** (66%)	**26** (63%)
Secondary to MDS, MPN, or prior chemo/radiotherapy	**17** (34%)	**15** (37%)
Cytogenetic Risk Group		
Intermediate	**27** (54%)	**23** (56%)
Cytogenetically normal	**21** (42%)	**17** (41%)
Cytogenetically abnormal	**6** (12%)	**6** (15%)
Poor*	**21** (42%)	**16** (39%)
Not classified	**2** (4%)	**2** (5%)

*Poor risk defined using SWOG definition, and included: Del(5q)/–5, –7/del(7q), abnormal 3q, 9q, 20q, 17p, t(6;9), t(9;22) and complex karyotypes (≥3 unrelated abnormalities)

- At baseline, 96 mutations in 28 genes were detected in 38 (93%) patients (**Figure 1**)
 - The most frequent mutations were in *SRSF2* (27%), *DNMT3A* (20%), *IDH2* (17%), *RUNX1* (17%), and *TET2* (17%)
 - The median number of mutations detected per patient was 2 (range: 0–6)
 - Distribution of mutations is in marked contrast with young *de novo* AML patients[1] and represents the unique features of elderly AML population
 - Frequent mutations that are related to secondary AML[5] and clonal hematopoiesis of indeterminate potential (CHIP)
 - The mutation profile appeared to be generally typical of an older population with AML[1] and are also common in myelodysplastic syndrome[6]
- Among the 33 patients with evaluable treatment response, CR was observed in 13 (39%) patients
 - The rate of CR was significantly higher in patients with mutations in *NPM1* or in one of the DNA methylation pathway genes
 - Patients with *TP53* mutation had a trend for poor CR (**Table 2**)

Figure 1. Landscape of High-Confidence Somatic Mutations Detected at Pre-Treatment Samples

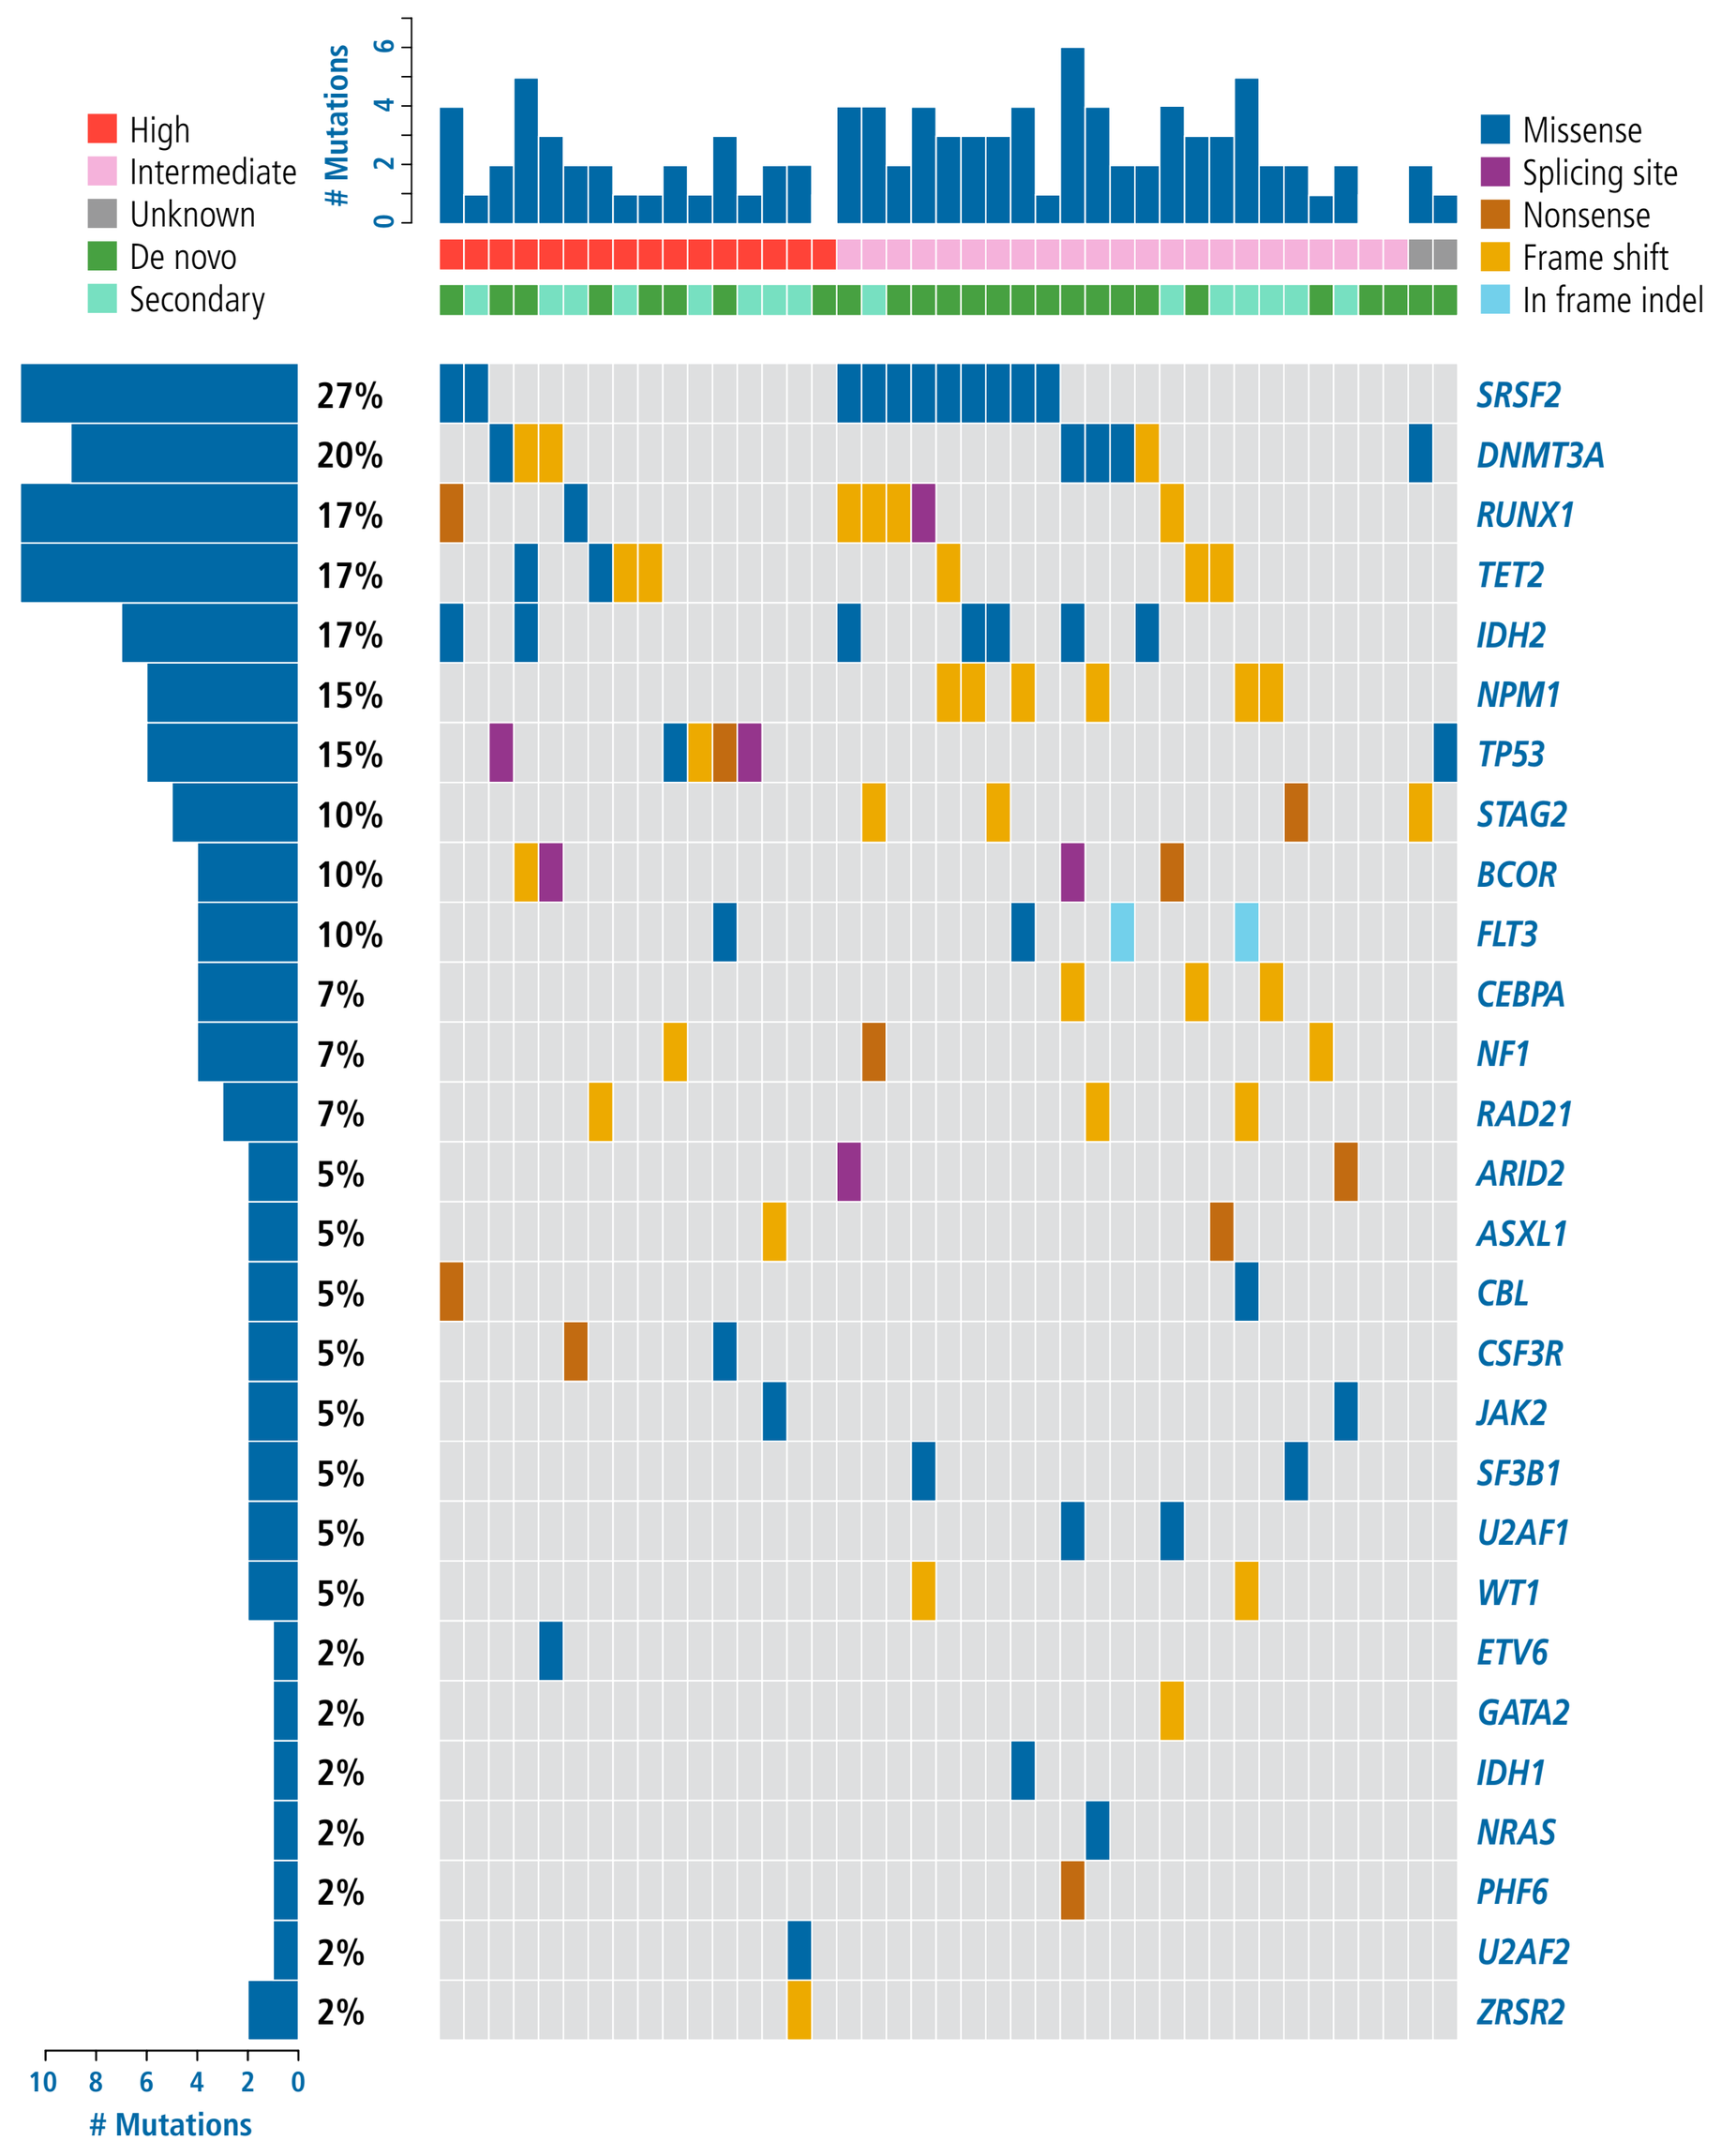
High
Intermediate
Unknown
De novo
Secondary
Mutations
Missense
Splicing site
Nonsense
Frame shift
In frame indel
27% SRSF2
20% DNMT3A
17% RUNX1
17% TET2
17% IDH2
15% NPM1
15% TP53
10% STAG2
10% BCOR
10% FLT3
7% CEBPA
7% NF1
7% RAD21
5% ARID2
5% ASXL1
5% CBL
5% CSF3R
5% JAK2
5% SF3B1
5% U2AF1
5% WT1
2% ETV6
2% GATA2
2% IDH1
2% NRAS
2% PHF6
2% U2AF2
2% ZRSR2
10 8 6 4 2 0
Mutations

Table 2. CR Rate by Mutation Status of Specific Genes*

Gene; n (%)	CR Rate (%) Mutated	Wild-type	*P* value
DNA methylation pathway†; 15 (37)	60	22	0.027
SRSF2; 11 (27)	22	46	0.204
RUNX1; 7 (17)	17	44	0.217
NPM1; 6 (15)	83	30	0.025
TP53; 5 (12)	0	46	0.065
BCOR; 4 (10)	75	35	0.157
FLT3; 4 (10)	67	37	0.338
STAG2; 4 (10)	0	45	0.118
CEBPA; 3 (7)	100	33	0.052
RAD21; 3 (7)	100	33	0.052
NF1; 3 (7)	0	41	0.606

*n≥3 patients ; †*DNMT3A*, *IDH1*, *IDH2*, or *TET2*

- Gene mutation associations with OS are shown in **Table 3** and **Figure 2**
 - The median follow-up duration was 23.8 months (95% CI: 20.4–27.1 months) with median OS of 18.1 months (95% CI: 10.1–26.1 months)
 - Median OS was 17.7 months in 20 patients with mutations typically associated with secondary AML[5] and 18.1 months in 18 patients with mutations typically associated with de novo AML[5]

Table 3. OS by Mutation Status of Specific Genes*

Gene; n (%)	Median OS (months) Mutated	Wild-type	*P* value
SRSF2; 11 (27)	10.7	19.1	0.230
DNMT3A; 8 (20)	13.5	19.1	0.874
IDH2; 7 (17)	Not reached	14.9	0.209
RUNX1; 7 (17)	8.0	18.2	0.078
NPM1; 6 (15)	Not reached	14.9	0.133
TET2; 5 (12)	20.1	17.8	0.786
TP53; 5 (12)	7.6	19.1	0.668
BCOR; 4 (10)	19.1	17.8	0.382
FLT3; 4 (10)	7.6	18.2	0.939
STAG2; 4 (10)	8.0	19.1	0.768
CEBPA; 3 (7)	Not reached	14.9	0.061
RAD21; 3 (7)	22.9	14.9	0.626
NF1; 3 (7)	3.0	19.1	0.005

*n≥3 patients

Figure 2. OS for Specific Genes With Mutations

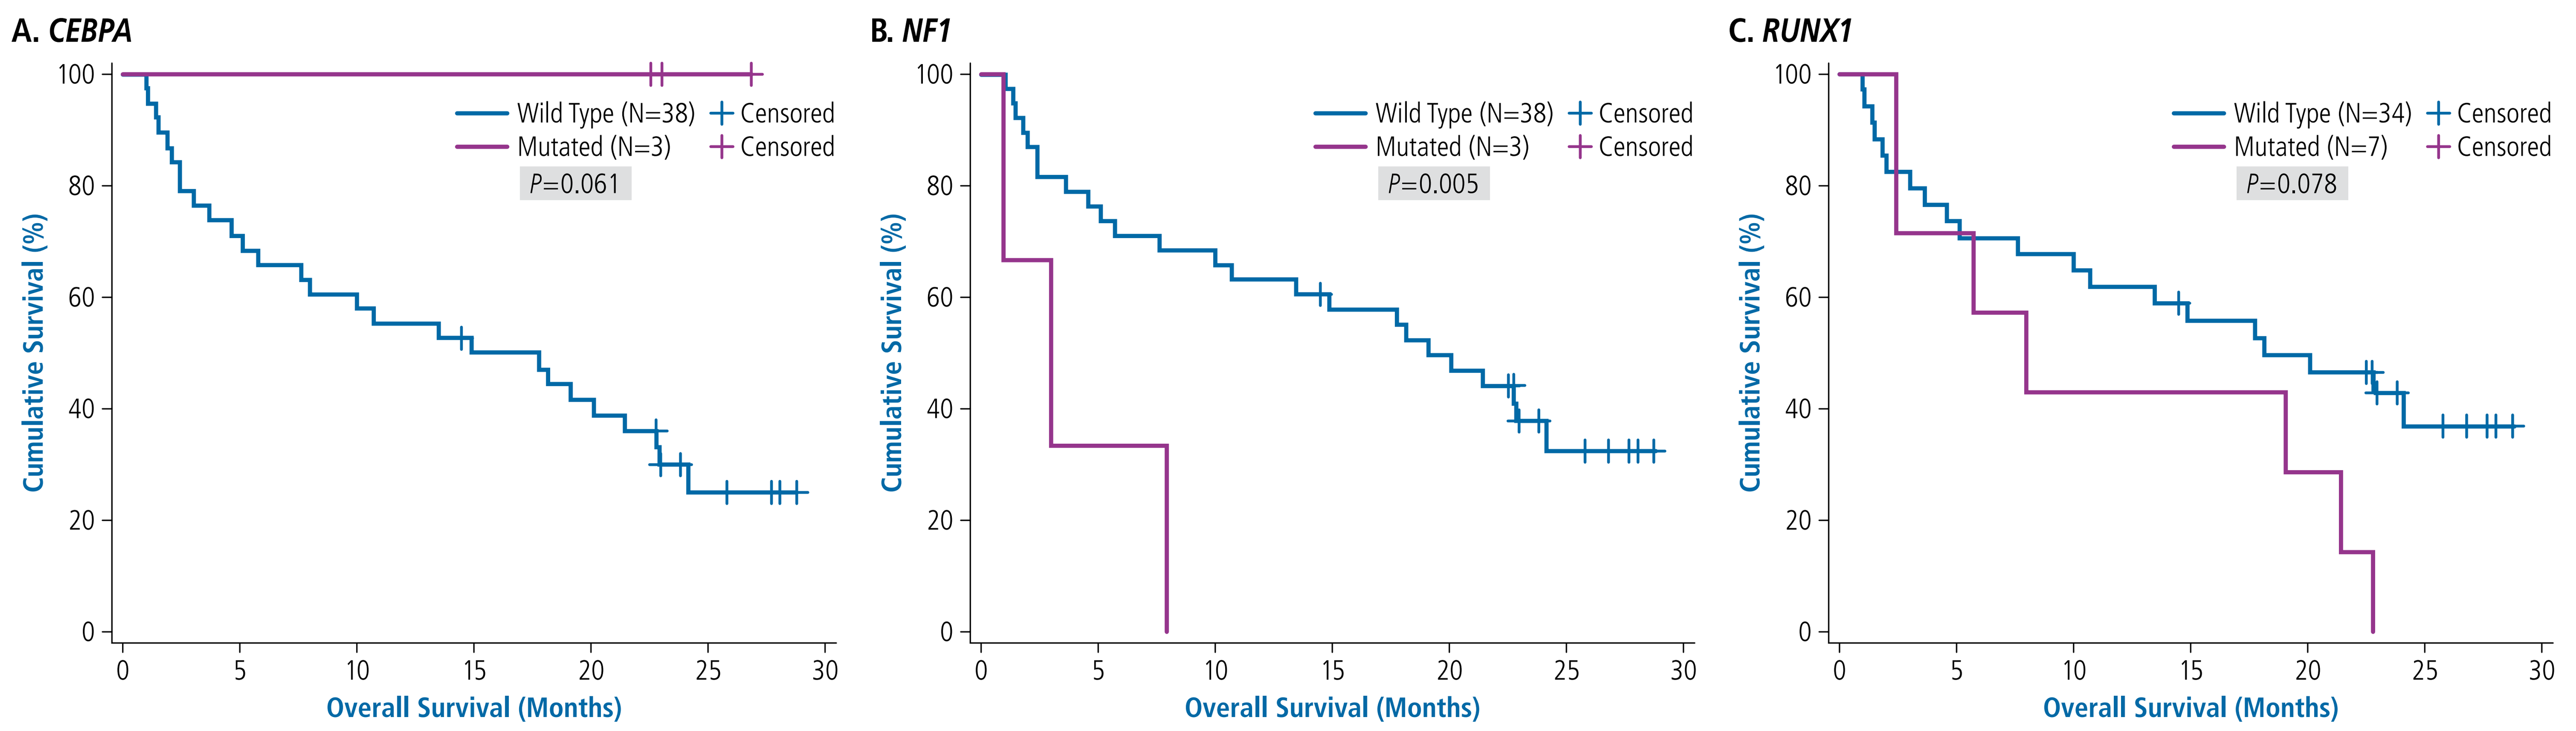
A. CEBPA
Wild Type (N=38) Censored
Mutated (N=3) Censored
P=0.061
B. NF1
Wild Type (N=38) Censored
Mutated (N=3) Censored
P=0.005
C. RUNX1
Wild Type (N=34) Censored
Mutated (N=7) Censored
P=0.078
Cumulative Survival (%)
Overall Survival (Months)

- A CR was achieved in 10/19 patients who had longitudinal sequencing analysis and at the time of CR, 9 (90%) had persistently detectable mutations in their bone marrow
 - In 7 of the 10 patients, continued exposure to pracinostat and azacitidine lowered the VAF or cleared residual mutations (**Figure 3A**)
 - Mutations in genes associated with DNA methylation, RNA splicing, clonal hematopoiesis of indeterminate potential (CHIP), and receptor tyrosine kinase (RTK) pathways had poor clearance of mutation
 - Pracinostat+azacitidine led to better clearance of transcription factors or cohesin
 - In 2 patients, relapsed samples were sequenced and showed re-expansion of the founder clone (**Figure 3B**)
 - Expansion of a new clone occurred in 1 patient (*RAD21*; **Figure 3C**)

Figure 3. Longitudinal Sequencing Analyses (Each Panel Shows Data for One Patient)

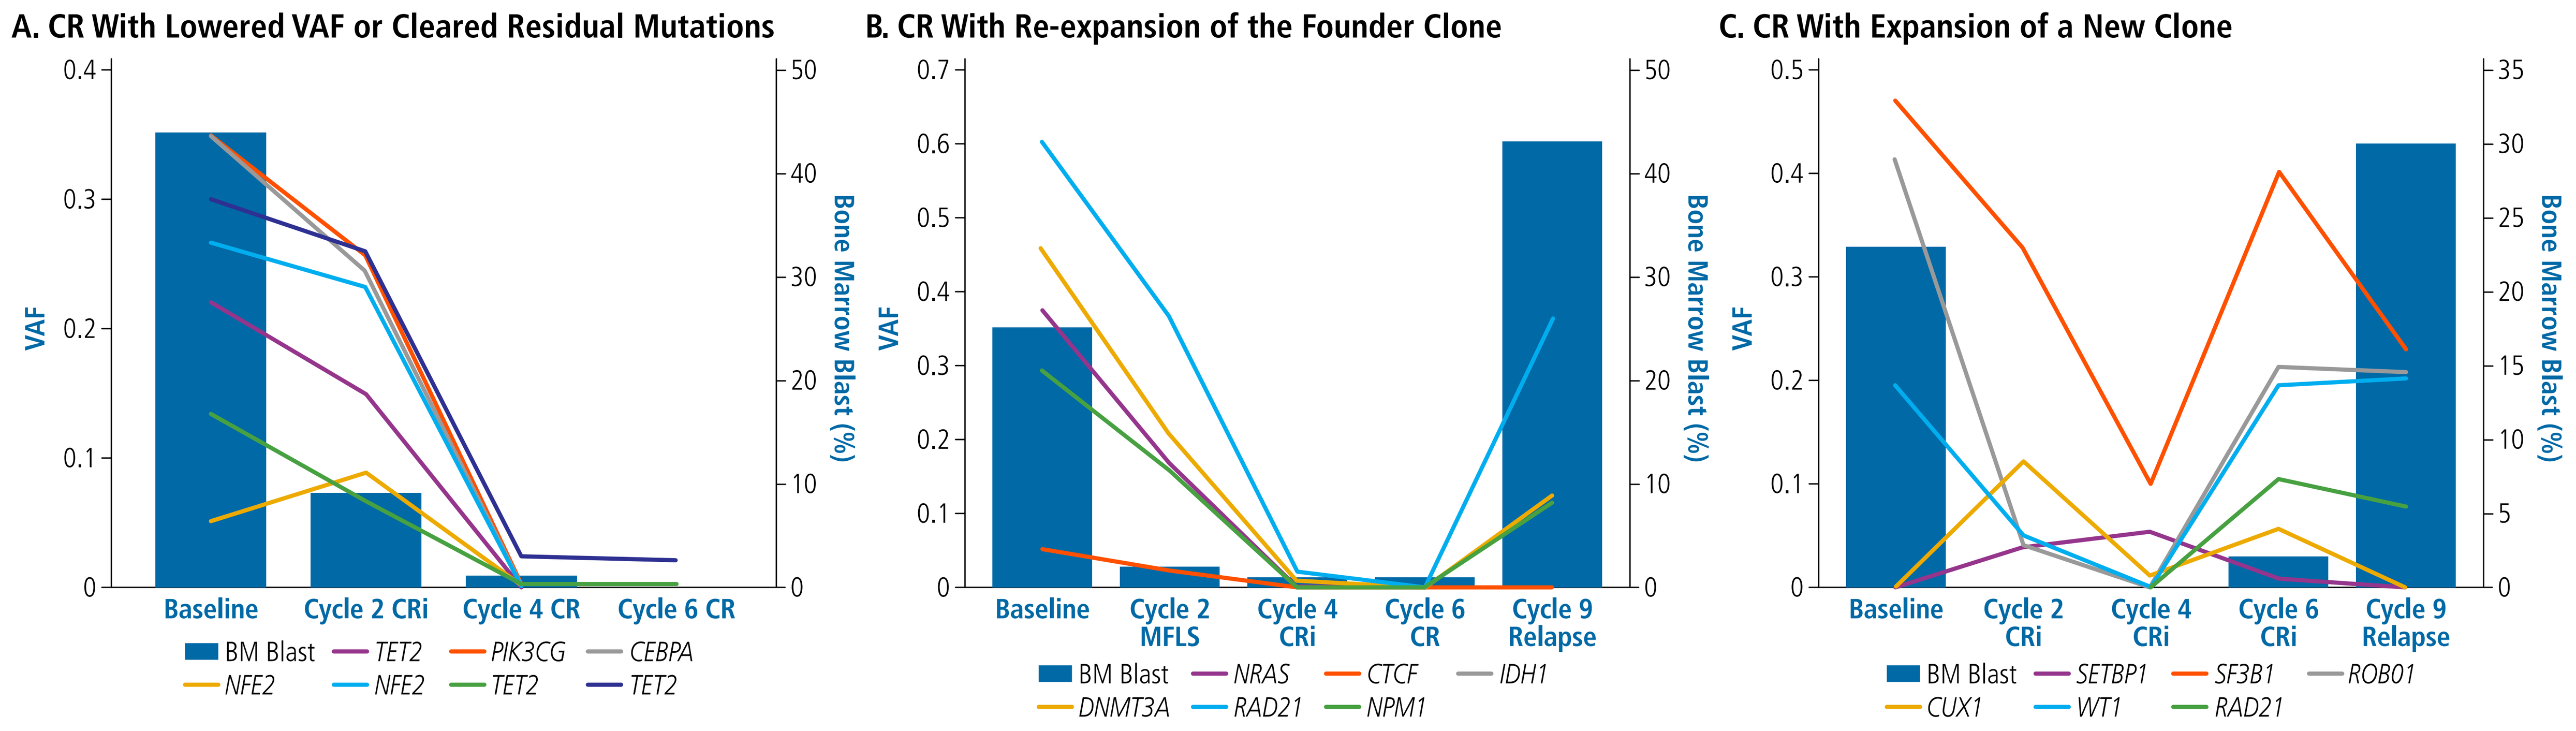
A. CR With Lowered VAF or Cleared Residual Mutations
VAF
Bone Marrow Blast (%)
Baseline
Cycle 2 CRi
Cycle 4 CR
Cycle 6 CR
BM Blast
TET2
PIK3CG
CEBPA
NFE2
NFE2
TET2
TET2
B. CR With Re-expansion of the Founder Clone
Baseline
Cycle 2 MFLS
Cycle 4 CRi
Cycle 6 CR
Cycle 9 Relapse
BM Blast
NRAS
CTCF
IDH1
DNMT3A
RAD21
NPM1
C. CR With Expansion of a New Clone
Baseline
Cycle 2 CRi
Cycle 4 CRi
Cycle 6 CRi
Cycle 9 Relapse
BM Blast
SETBP1
SF3B1
ROB01
CUX1
WT1
RAD21

CONCLUSIONS

- Mutations in *NPM1* and the DNA methylation pathway were common in the study population and associated with a better response to pracinostat and azacitidine, while *TP53* mutation was associated with a poor response
 - Mutations that respond to pracinostat and azacitidine may be an important factor in the favorable CR and OS findings in the overall study
- Persistent mutation at the time of CR suggests residual leukemic clonal hematopoiesis
- Continued treatment increases the rate of minimal residual disease clearance as by continued decline of mutation VAF after achieving CR

REFERENCES: **1.** Döhner H, Weisdorf DJ, Bloomfield CD. Acute myeloid leukemia. *N Engl J Med.* 2015;373(12):1136–1152. **2.** Dombret H, Seymour JF, Butrym A, et al. International phase 3 study of azacitidine vs conventional care regimens in older patients with newly diagnosed AML with >30% blasts. *Blood.* 2015;126(3):291–299. **3.** Schneider-Stock R, Ocker M. Epigenetic therapy in cancer: molecular background and clinical development of histone deacetylase and DNA methyltransferase inhibitors. *IDrugs.* 2007;10(8):557–561. **4.** Garcia-Manero G, Atallah E, Khaled SK, et al. A phase 2 study of pracinostat and azacytidine in elderly patients with acute myeloid leukemia (AML) not eligible for induction chemotherapy: response and long-term survival benefit. *Blood* (Annual Meeting Abstracts). 2016;128(22):100. **5.** Lindsley RC, Mar BG, Mazzola E, et al. Acute myeloid leukemia ontogeny is defined by distinct somatic mutations. *Blood.* 2015;125(9):1367–1376. **6.** Lindsley RC, Saber W, Mar BG, et al. Prognostic mutations in myelodysplastic syndrome after stem-cell transplantation. *N Engl J Med.* 2017;376(6):536–547.

DISCLOSURES: Koichi Takahashi, Yasmin Abaza, Feng Wang, Curtis Gumbs, Song Xingzhi, Andrew Futreal, Bruno C. Medeiros, Samer K. Khaled, Mrinal M. Patnaik, and Guillermo Garcia-Manero have nothing to disclose. Ehab Atallah has received research funding from Takeda and honoraria from Ariad; has served as a consultant for BMS, Novartis, CIT Biopharma, and Incyte; and has served as a grant reviewer for Pfizer. Martha Arellano has received research funding from Cephalon. Elena Palmesino is an employee of Helsinn Healthcare.

SUPPORT: The clinical study was supported by MEI and the molecular study by Helsinn. Helsinn Healthcare, SA provided funding to The Curry Rockefeller Group, LLC, for support in writing and editing this poster.